Knowledge-driven drug development: the quest for a semantic repository of clinical studies

Kaushal Desai, AstraZeneca Pharmaceuticals

SCIENTIFIC RATIONALE

- Knowledge exploitation drives innovation in drug development.
- Knowledge generated in clinical studies is often stored in disconnected 'silos' in structured databases as well as 'unstructured' study documents.
- Challenges associated with **access and re-use** of knowledge therefore, limit cross-compound, cross-study information expoitation.
- A semantic repository of clinical studies could provide the capability to –
 - Respond efficiently to regulatory queries and avoid excessive, repititive integration effort .
 - Enable information exploitation through greater control over and utilization of organization knowledge.
 - Deliver automated reasoning capability to proactively identify optimal study designs based on successful historic outcomes.

CONCEPTUAL SOLUTION

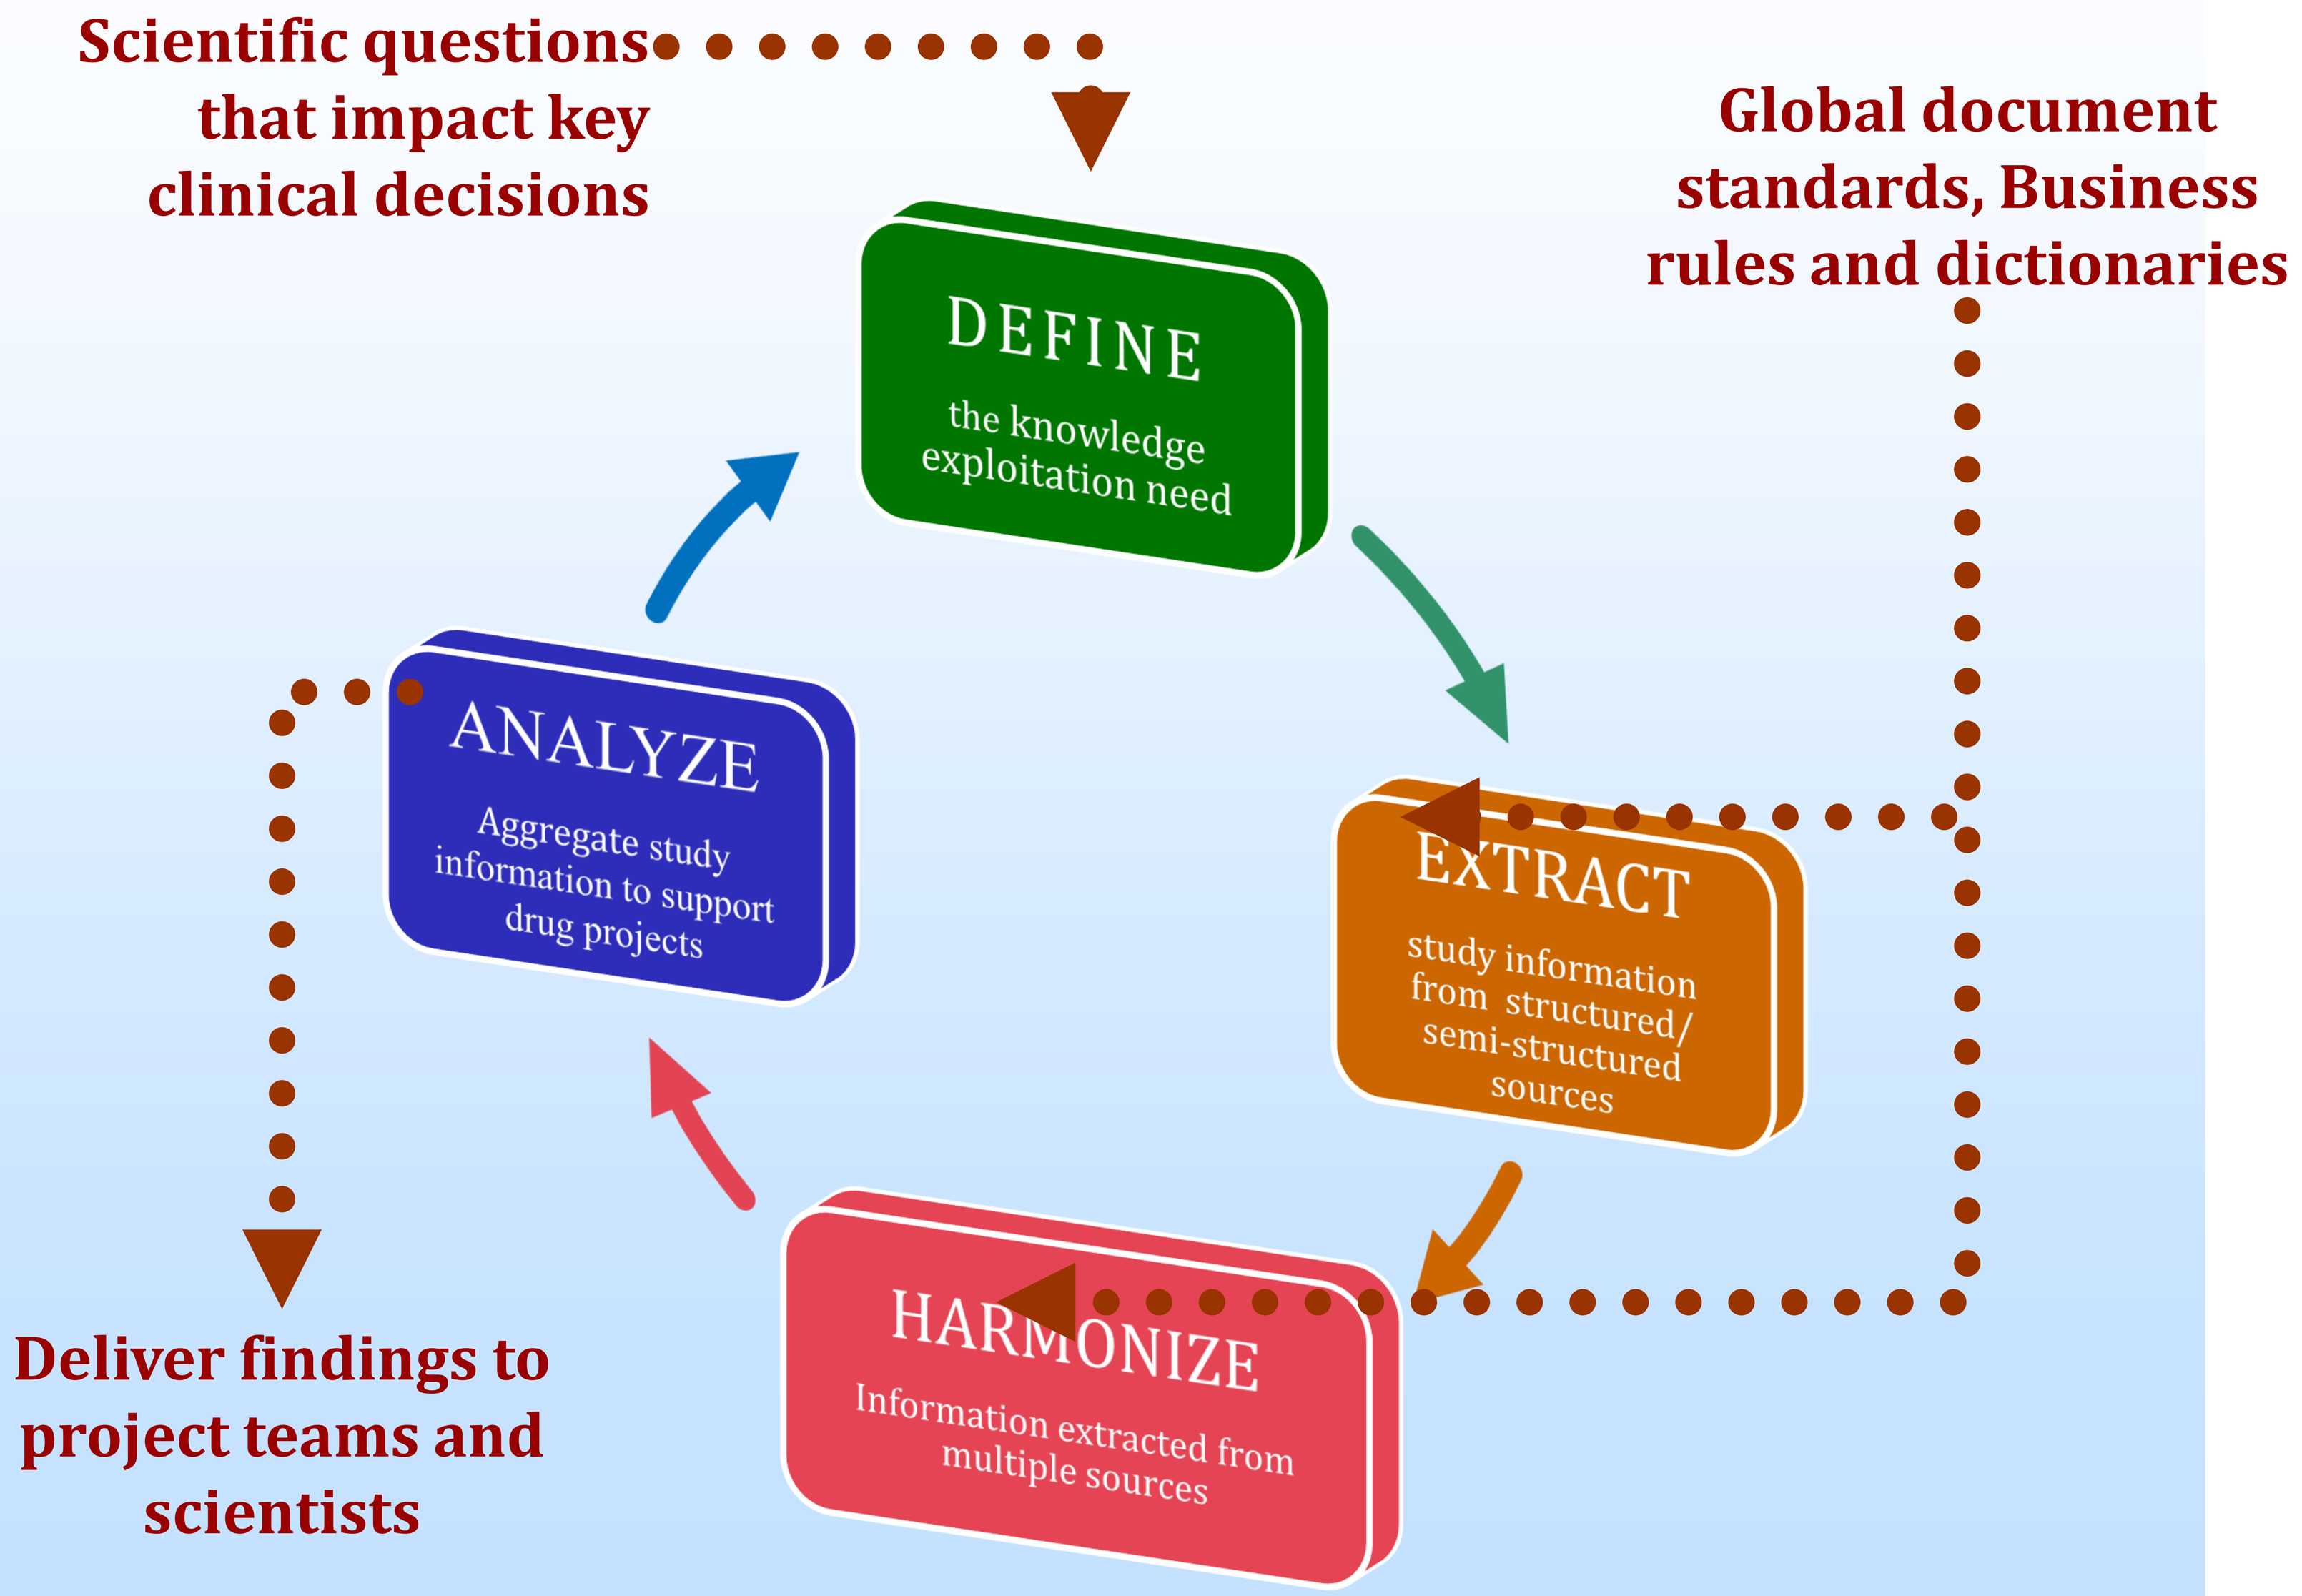
Scientific questions that impact key clinical decisions
Global document standards, Business rules and dictionaries
DEFINE
the knowledge exploitation need
EXTRACT
study information from structured/ semi-structured sources
HARMONIZE
Information extracted from multiple sources
ANALYZE
Aggregate study information to support drug projects
Deliver findings to project teams and scientists

SEMANTIC ANNOTATION OF CLINICAL STUDIES

Deliverables

- The ability to search clinical studies across multiple underlying databases and document stores, based on **planning** (study objectives, study design etc.), **execution** (milestone dates, events, number of subjects etc.) and **outcome** (adverse events, efficacy, PK results etc.) related criteria.

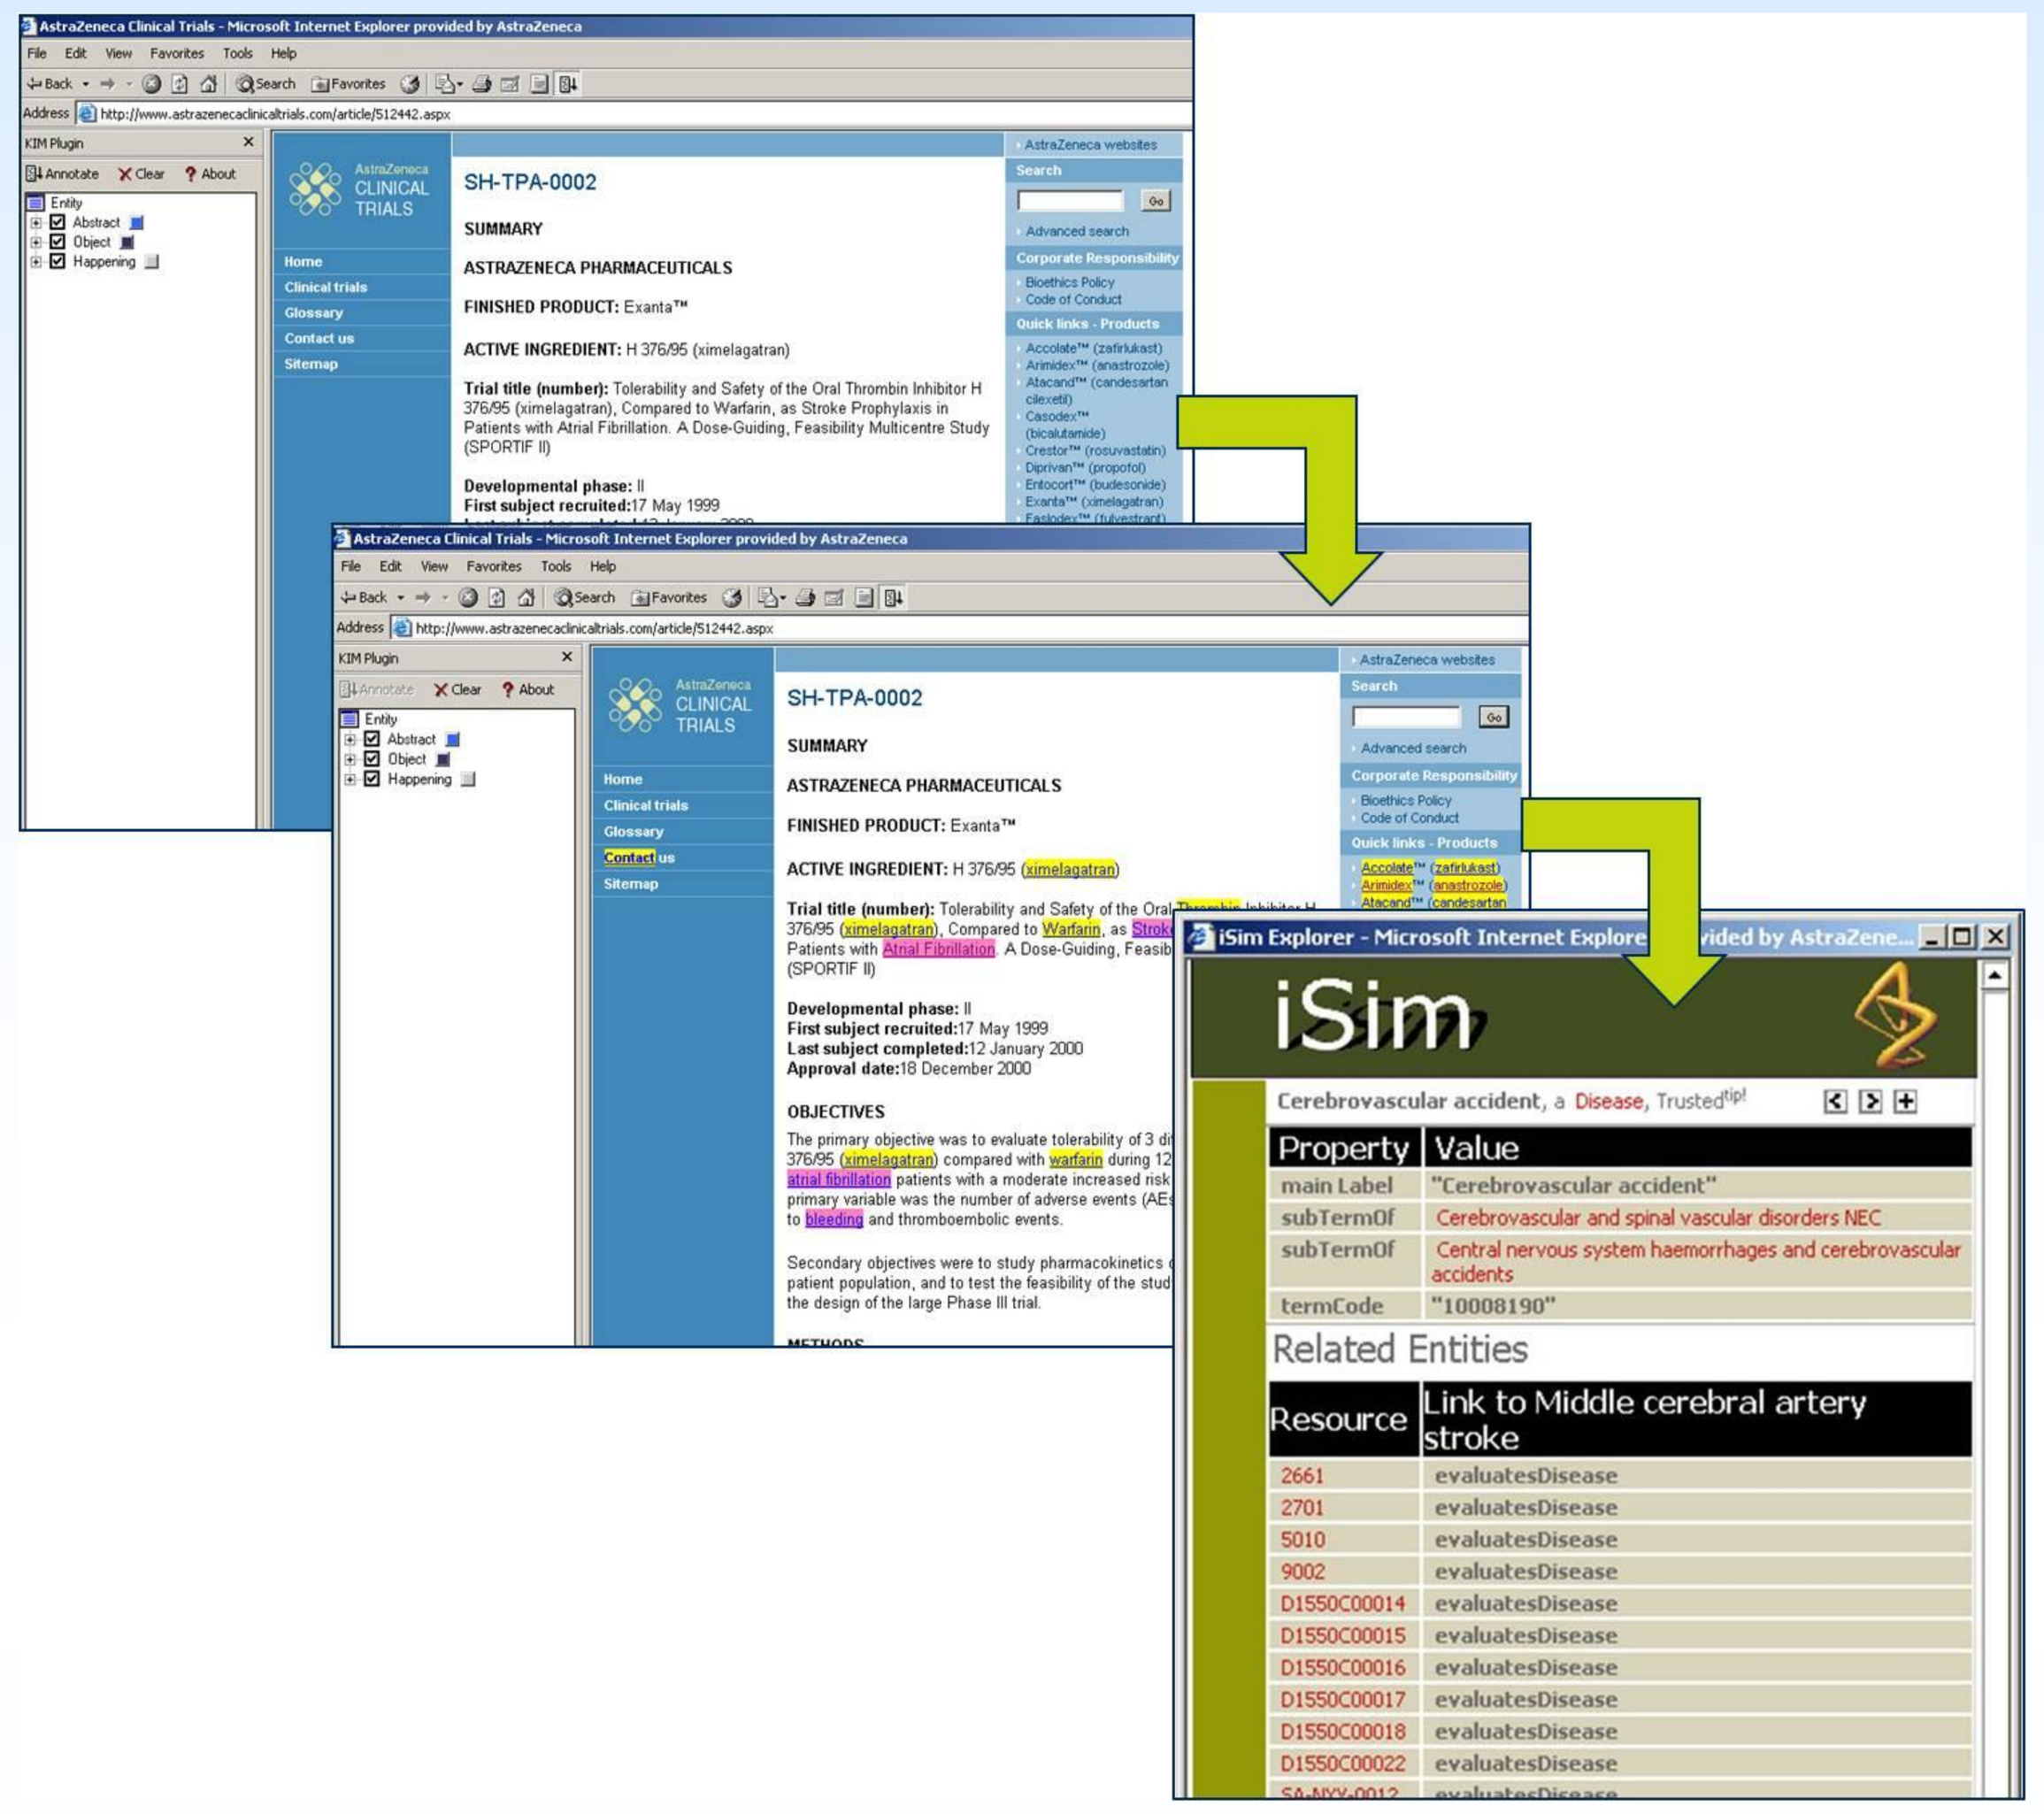
SH-TPA-0002
iSim
Property
Value
Related Entities
Resource
Link to Middle cerebral artery stroke

RDF/OWL FRAMEWORK FOR INTEGRATION

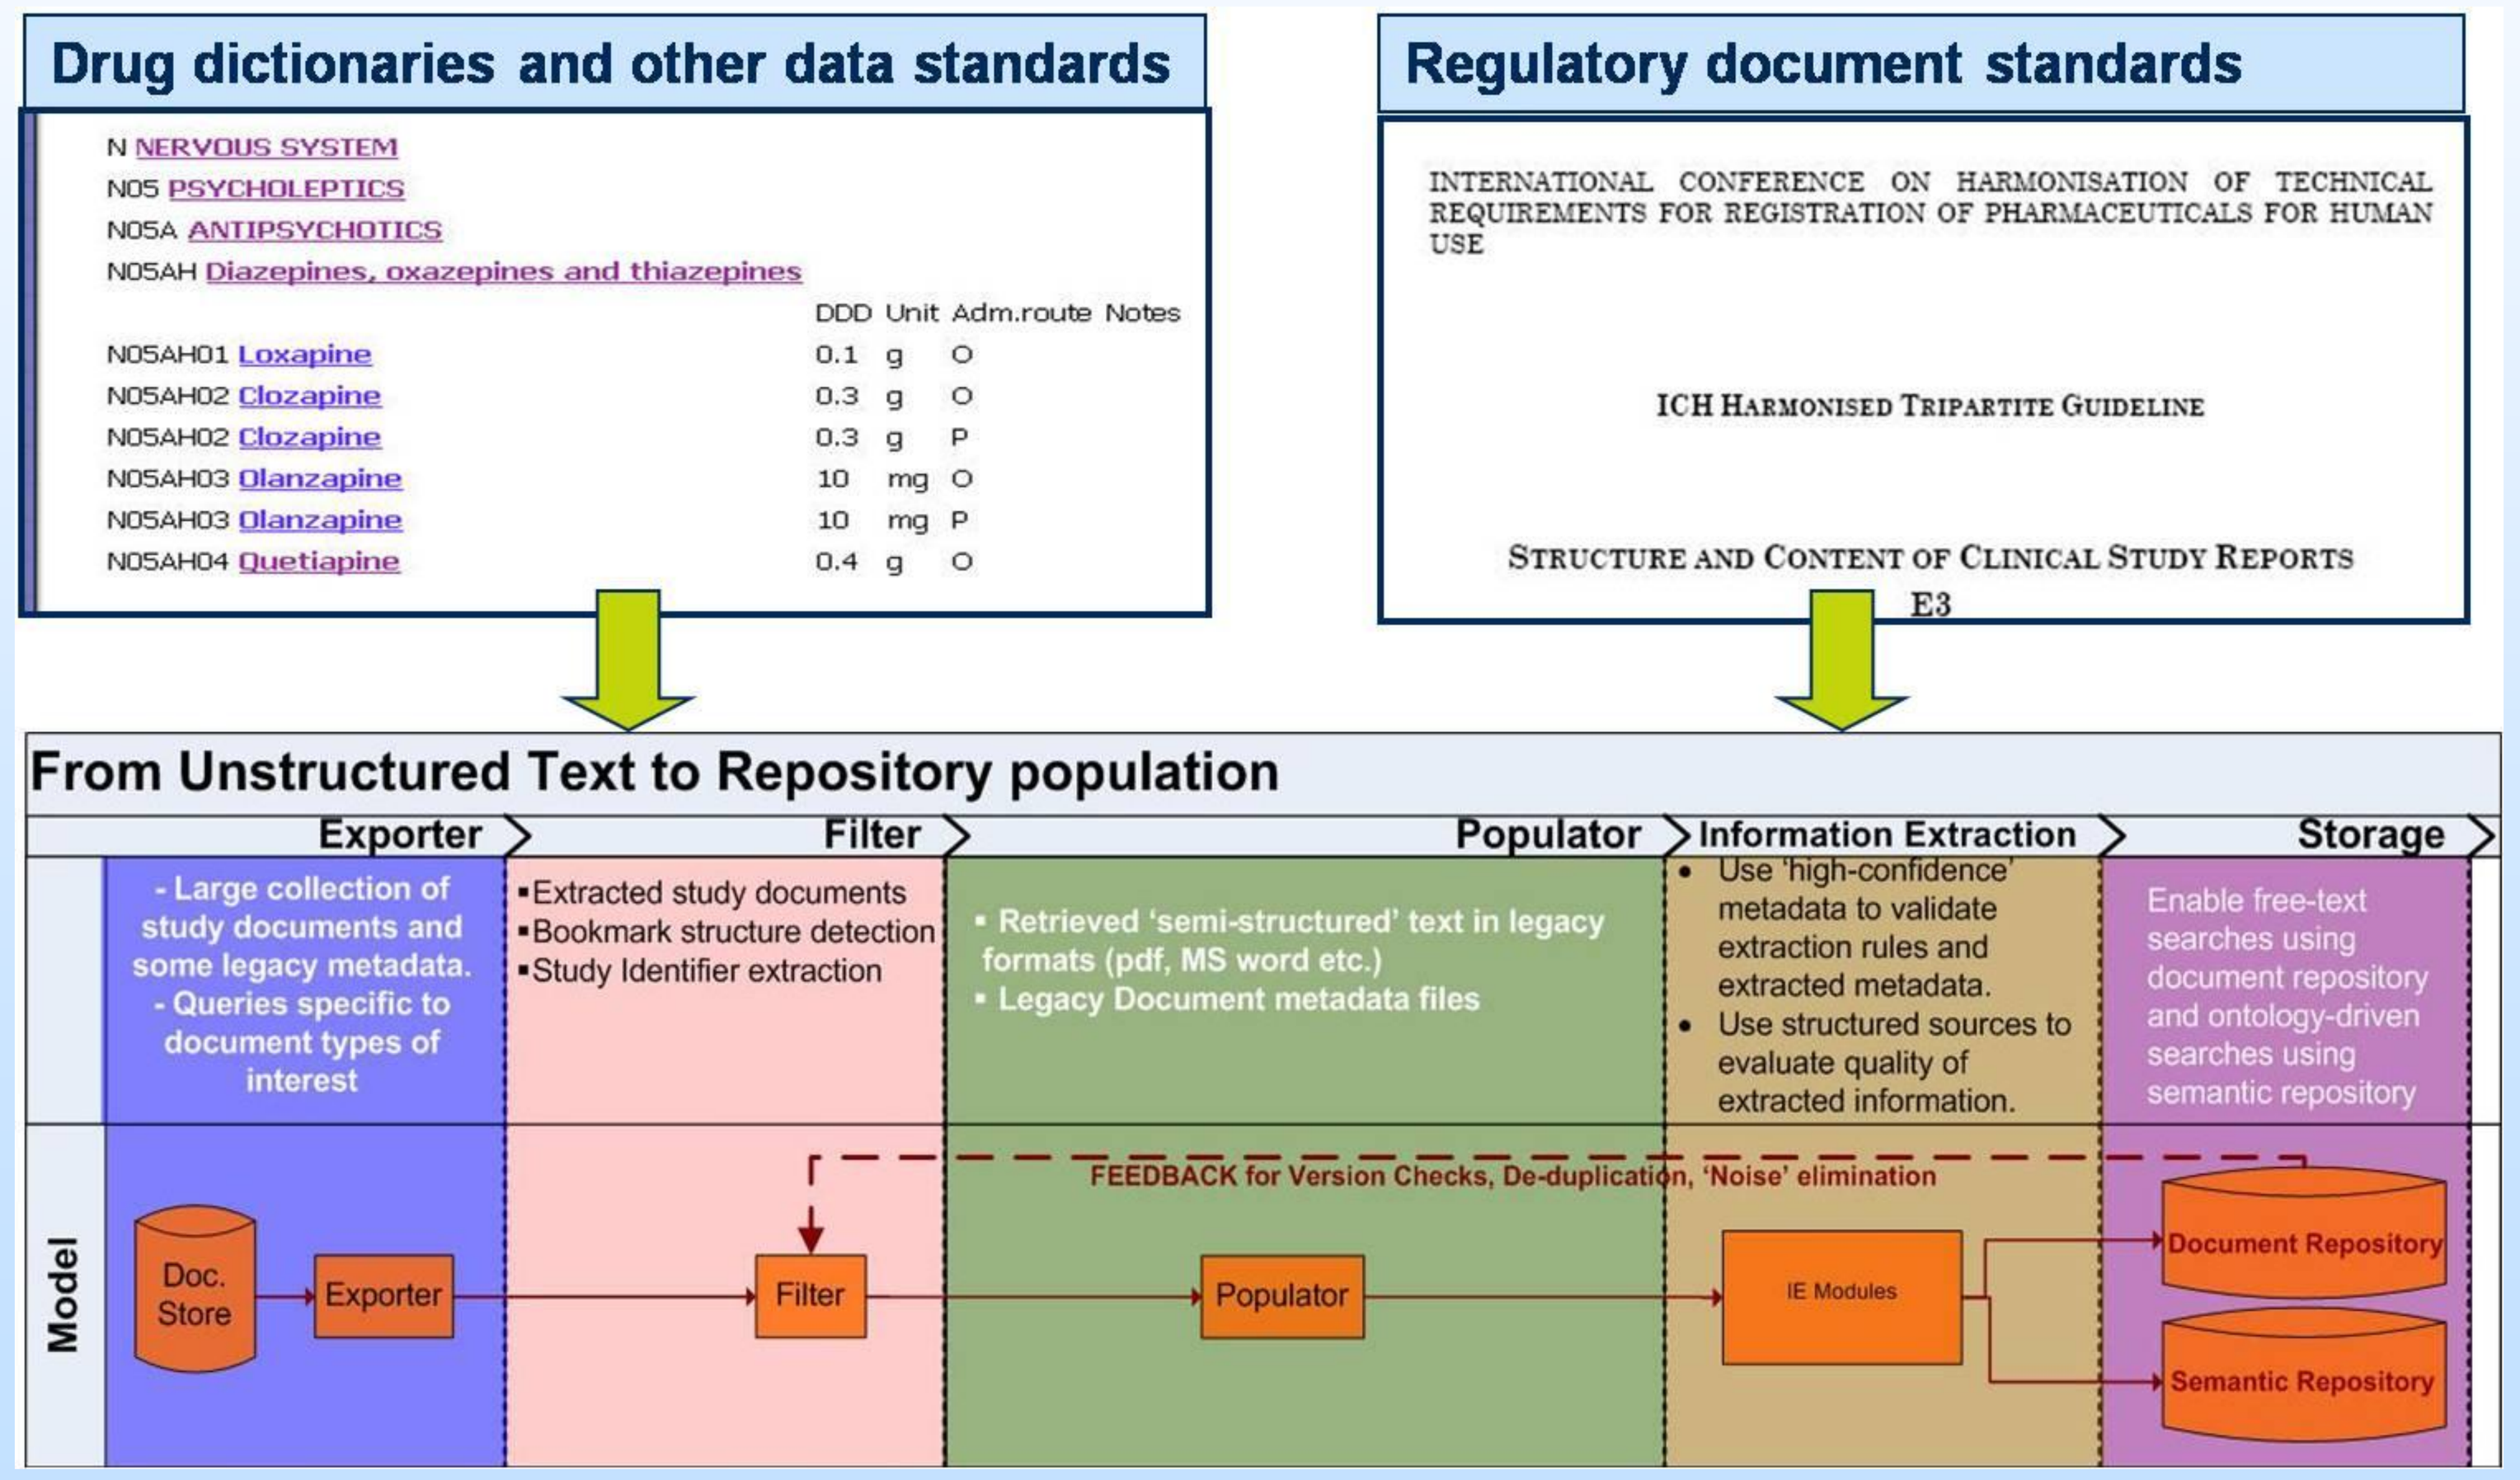
Drug dictionaries and other data standards
N NERVOUS SYSTEM
N05 PSYCHOLEPTICS
N05A ANTIPSYCHOTICS
N05AH Diazepines, oxazepines and thiazepines
DDD Unit Adm.route Notes
N05AH01 Loxapine 0.1 g O
N05AH02 Clozapine 0.3 g O
N05AH02 Clozapine 0.3 g P
N05AH03 Olanzapine 10 mg O
N05AH03 Olanzapine 10 mg P
N05AH04 Quetiapine 0.4 g O
Regulatory document standards
INTERNATIONAL CONFERENCE ON HARMONISATION OF TECHNICAL REQUIREMENTS FOR REGISTRATION OF PHARMACEUTICALS FOR HUMAN USE
ICH HARMONISED TRIPARTITE GUIDELINE
STRUCTURE AND CONTENT OF CLINICAL STUDY REPORTS
E3
From Unstructured Text to Repository population
Exporter
Filter
Populator
Information Extraction
Storage
- Large collection of study documents and some legacy metadata.
- Queries specific to document types of interest
▪Extracted study documents
▪Bookmark structure detection
▪Study Identifier extraction
• Retrieved 'semi-structured' text in legacy formats (pdf, MS word etc.)
• Legacy Document metadata files
• Use 'high-confidence' metadata to validate extraction rules and extracted metadata.
• Use structured sources to evaluate quality of extracted information.
Enable free-text searches using document repository and ontology-driven searches using semantic repository
Model
FEEDBACK for Version Checks, De-duplication, 'Noise' elimination
Doc. Store
Exporter
Filter
Populator
IE Modules
Document Repository
Semantic Repository

BENEFITS FROM THE SEMANTIC APPROACH

- **How well do semantic tools enable the utilization of information and knowledge ?**
 - Within 1.5 years of deployment, the information integration benefits resulted in more than 7 years of work savings on various drug project and information exploitation teams.
 - Benefits from reasoning on clinical study information are yet to be realized.
- **What are the measurable benefits of using an RDF/OWL framework ?**
 - Abstraction from drug-related 'facts' to drug-classes and abstraction from adverse event-related 'facts' to System organ class would be cumbersome in case of a 'relational' architecture.
 - Integrated search on studies in a global organization without forcing a single 'static' information model on source systems would be difficult with a 'relational' solution.
 - Future scalability and addition of 'new knowledge' would be difficult without use of uniformly accepted standards.

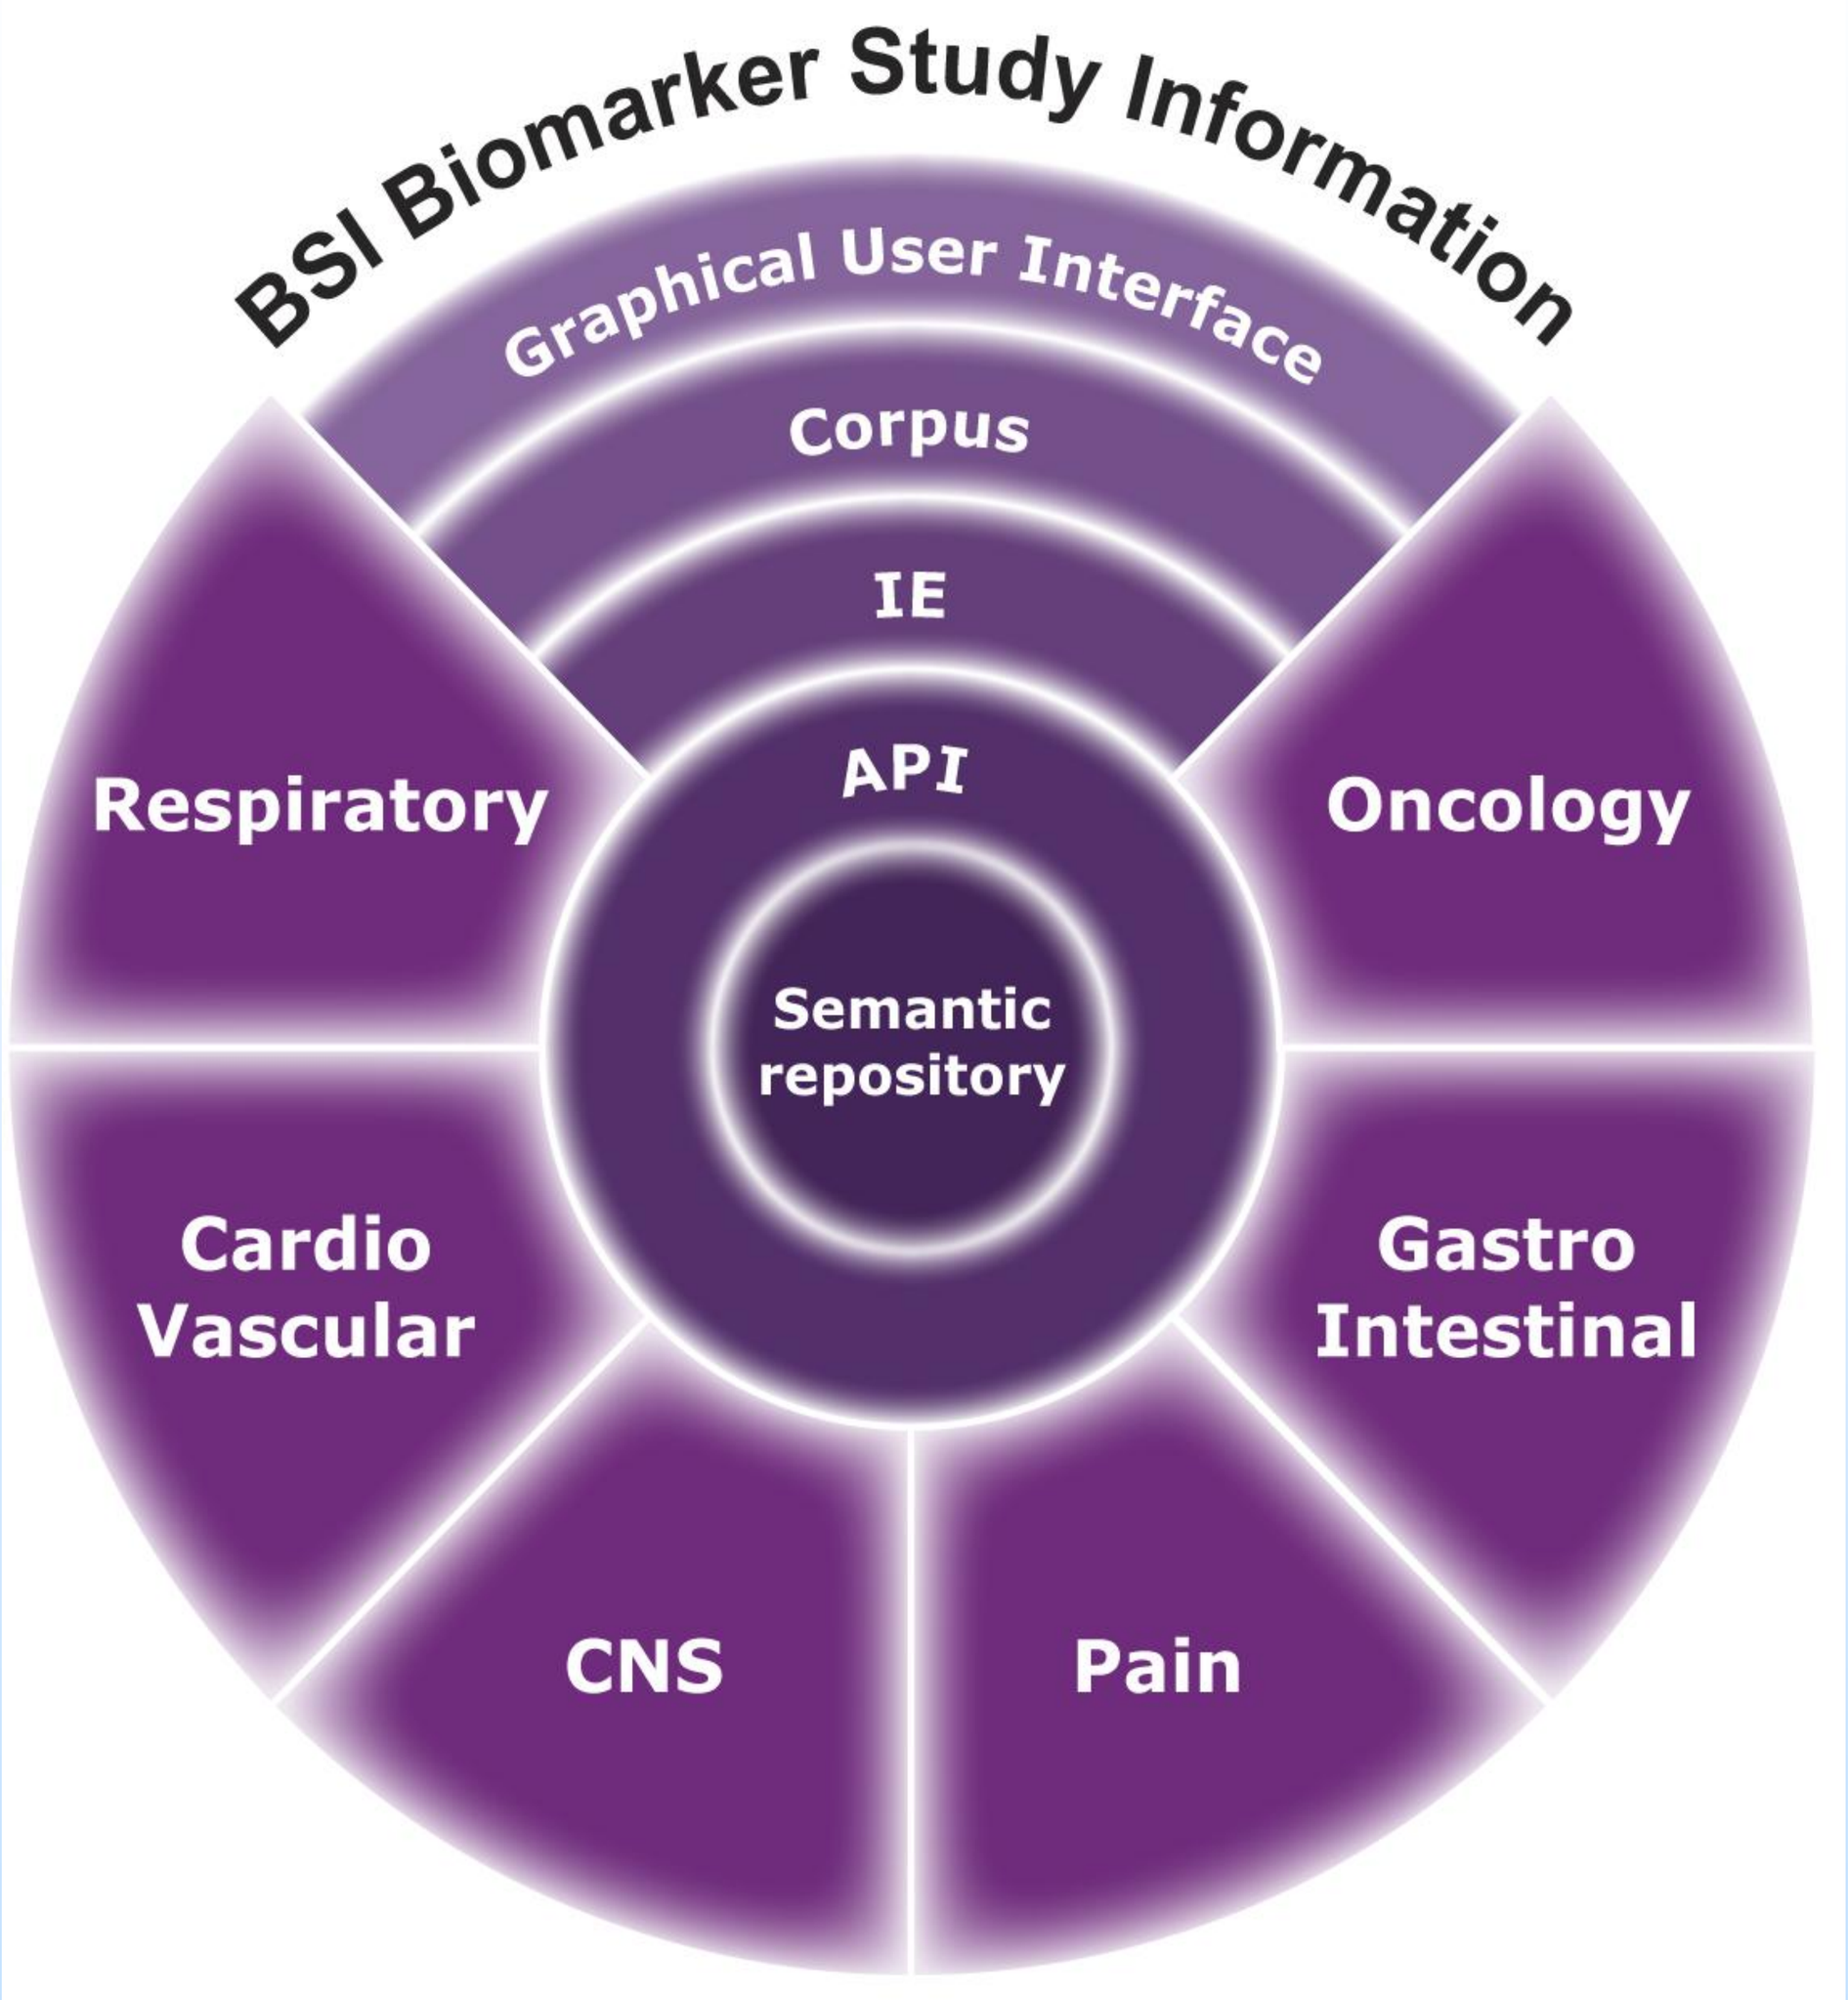
BSI Biomarker Study Information
Graphical User Interface
Corpus
IE
API
Semantic repository
Respiratory
Oncology
Cardio Vascular
Gastro Intestinal
CNS
Pain

REFERENCES

- Kaushal Desai, Borislav Popov. *Semantic annotation of clinical studies for knowledge-driven drug development*. European Semantic Technology conference, Austria, Vienna 2008.
- McCallum A., *Information Extraction: Distilling Structured data from unstructured text*. ACM Queue, November 2005.
- Gardner SP. *Ontologies and semantic data integration*. DDT, july 2005.

ACKNOWLEDGEMENTS

- **Business Team:** Kaushal Desai (Project lead), James Weatherall, Bosse Andersson.
- **IS Collaborators: E**lisabet Soderheilm (project manager), Frederik Lundbeck (technical lead), Bob Keesler, Tobias Heed, Mattias Almen).

External Partner

- **Ontotext Labs (Sirma Group)**

 KIM (Knowledge and Information Management) platform is a software platform for automatic ontology population and open-domain dynamic semantic annotation of unstructured and semi-structured content for Semantic Web and knowledge management applications.